Analytics 2.0: Unveiling Regional Care Gaps Through Innovative Educational Needs Assessment

Lucia Kim; Dustin Ensign, BSc, CHCP

Background & Objectives

Regional care variation presents a significant challenge in healthcare delivery, particularly in endocrinology and mental health.[1,2] This project analyzed the relationship between clinician interest and knowledge/confidence levels to inform targeted educational strategies across US regions, focusing on Type 2 Diabetes Mellitus (T2DM), obesity, and Major Depressive Disorder (MDD).[1,2,3] Through analysis of Pri-Med educational assessment data and regional care metrics,[4] the research developed actionable recommendations for healthcare systems and/or education providers to implement strategic initiatives that address regional care variation in these clinical areas. This project ultimately aims to provide insights that can guide the development of targeted educational programs to help reduce care disparities across different geographic regions.

Methods

We conducted a retrospective analysis of baseline knowledge and confidence scores from 2022-2023 Pri-Med educational programs (N=7,922), integrated with 2023-2024 Pri-Med clinician interest survey data (N=742). The analysis focused on regional variations in T2DM, obesity, and mental health education outcomes, segmented by major US regions and states.

High-Level Results

Our analysis revealed significant regional variations in both knowledge levels and clinician confidence. The Southwest region demonstrated the highest correct response rates (67%) for assessment questions about atypical antidepressants, while the Northeast showed the lowest (52%). For T2DM management, the Southeast region demonstrated the highest mastery (13%) for injectable semaglutide protocols, while other regions showed varying levels of competency. The Mid-Atlantic region led in obesity management knowledge, with 71% correctly identifying motivational interviewing objectives.

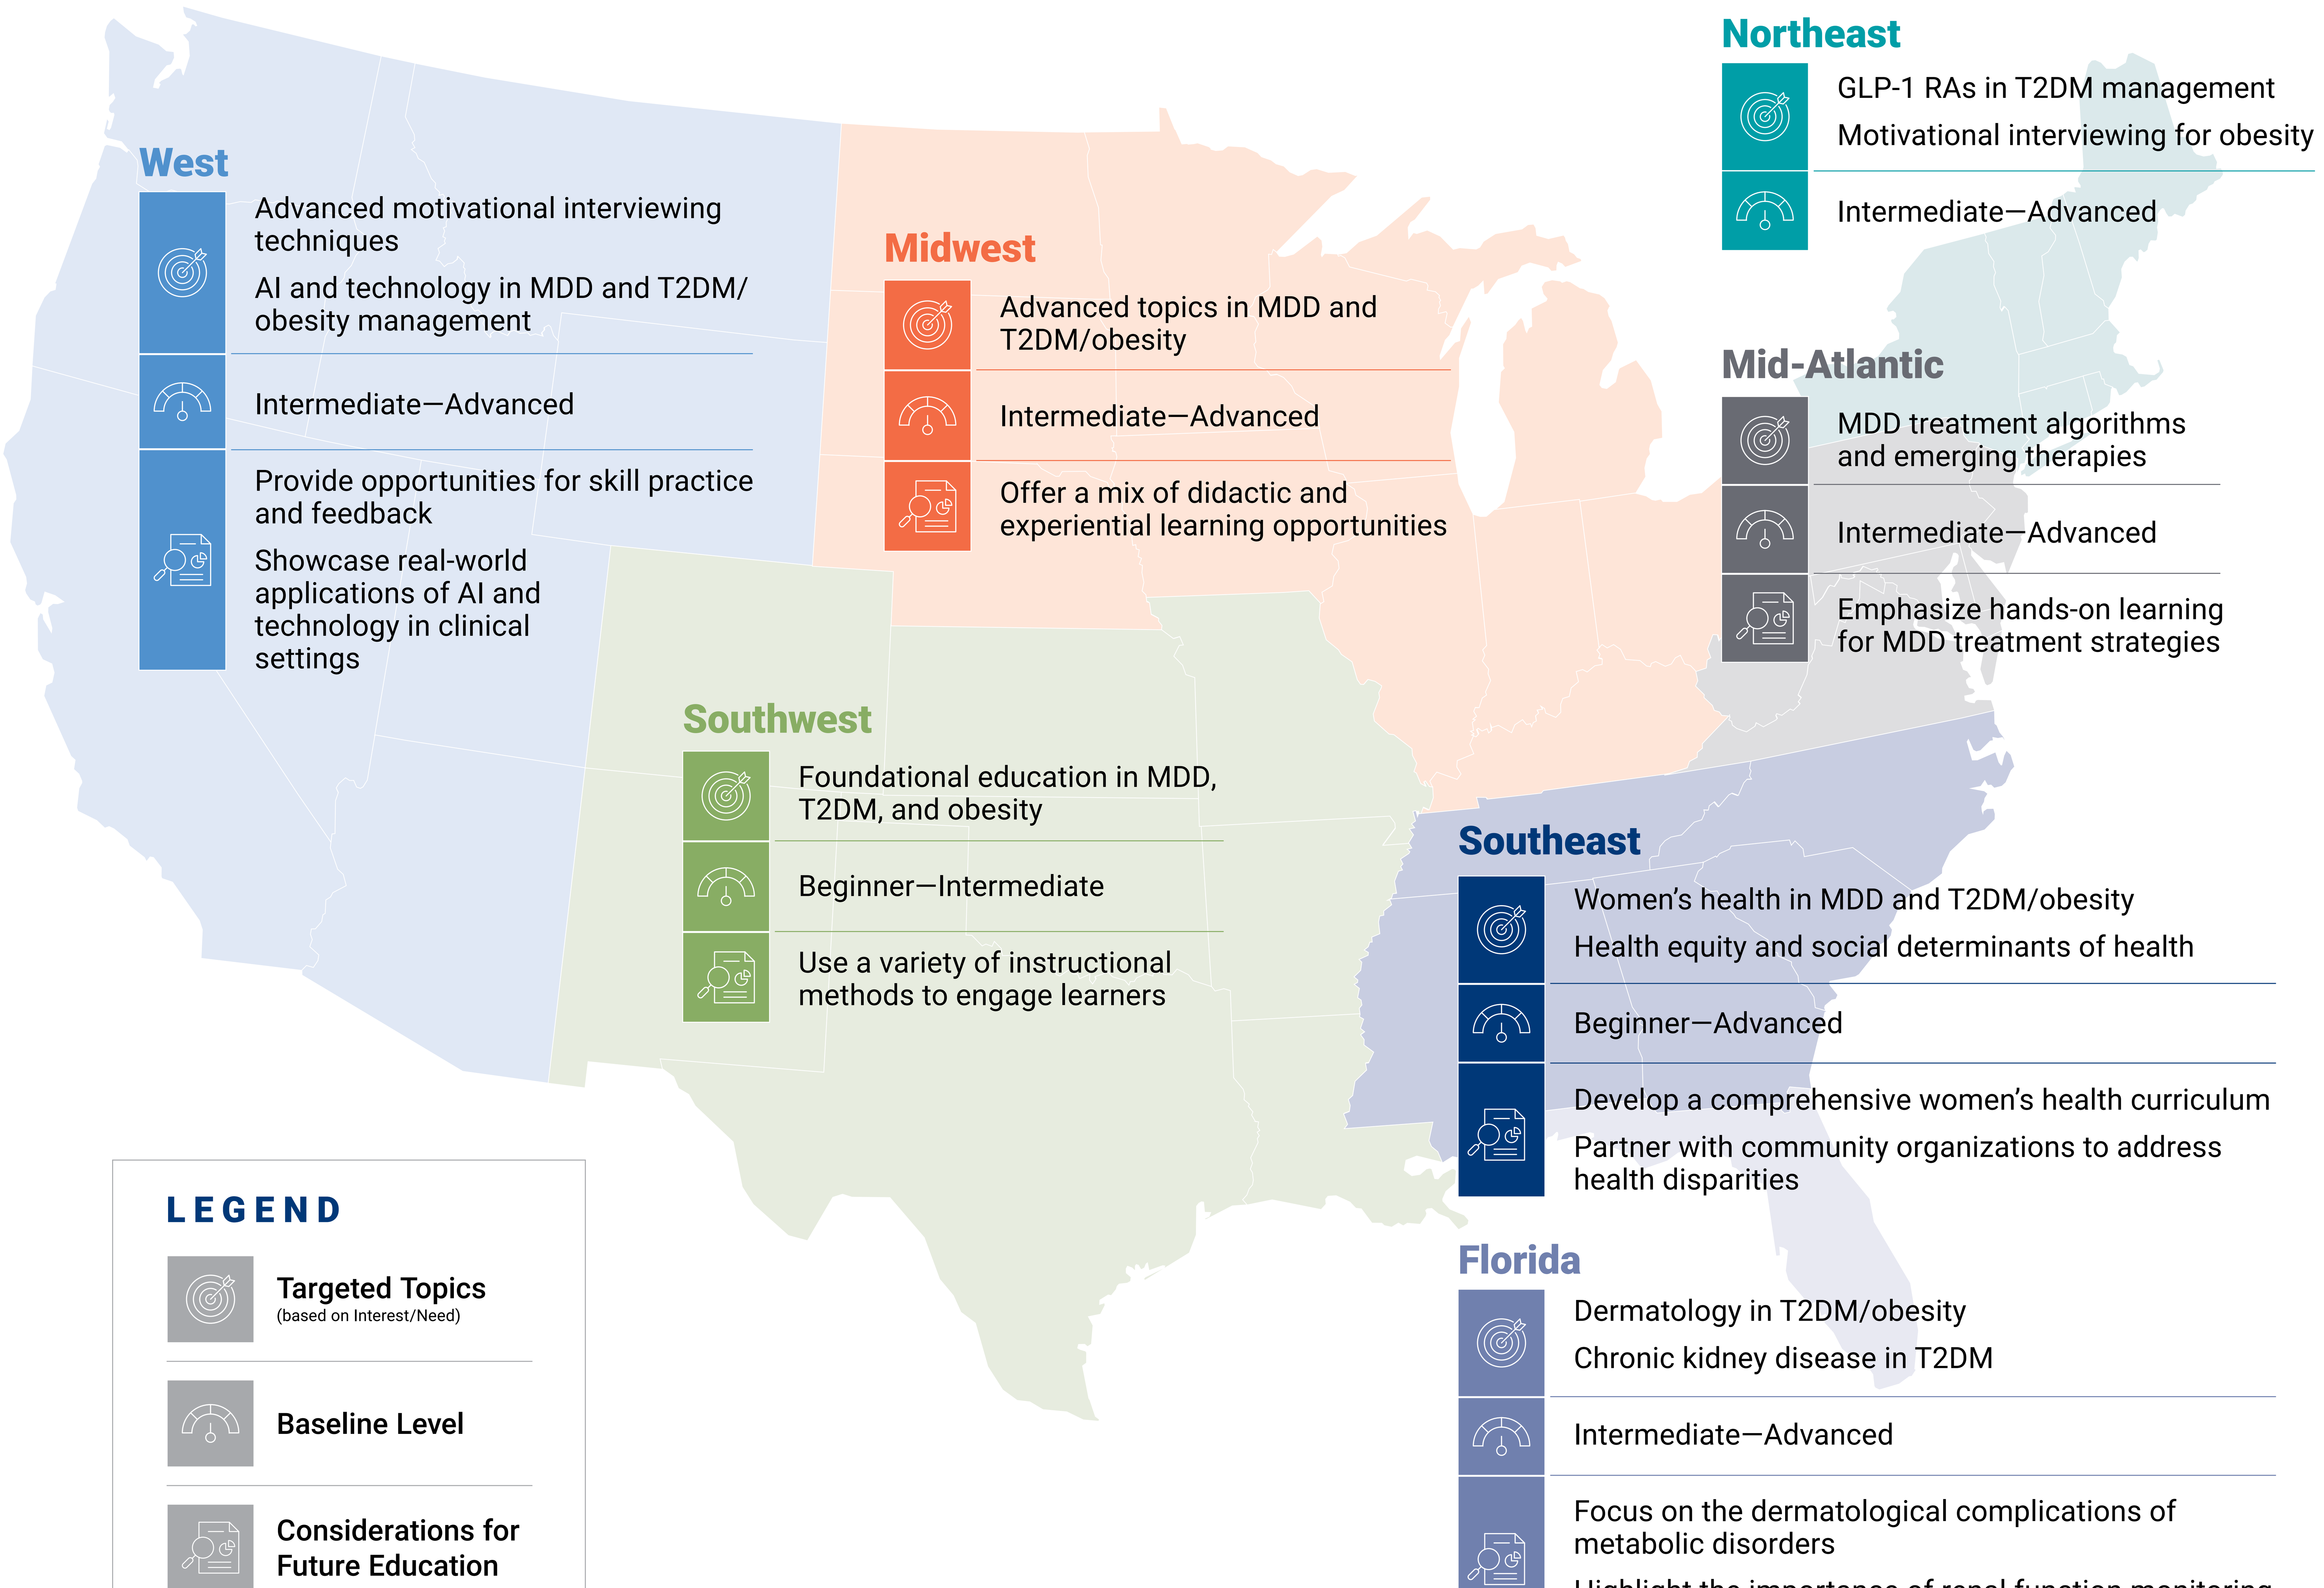

West

- Targeted topics: Advanced motivational interviewing techniques; AI and technology in MDD and T2DM/obesity management
- Baseline level: Intermediate—Advanced
- Considerations for future education: Provide opportunities for skill practice and feedback; Showcase real-world applications of AI and technology in clinical settings

Midwest

- Targeted topics: Advanced topics in MDD and T2DM/obesity
- Baseline level: Intermediate—Advanced
- Considerations for future education: Offer a mix of didactic and experiential learning opportunities

Northeast

- Targeted topics: GLP-1 RAs in T2DM management; Motivational interviewing for obesity
- Baseline level: Intermediate—Advanced

Mid-Atlantic

- Targeted topics: MDD treatment algorithms and emerging therapies
- Baseline level: Intermediate—Advanced
- Considerations for future education: Emphasize hands-on learning for MDD treatment strategies

Southwest

- Targeted topics: Foundational education in MDD, T2DM, and obesity
- Baseline level: Beginner—Intermediate
- Considerations for future education: Use a variety of instructional methods to engage learners

Southeast

- Targeted topics: Women's health in MDD and T2DM/obesity; Health equity and social determinants of health
- Baseline level: Beginner—Advanced
- Considerations for future education: Develop a comprehensive women's health curriculum; Partner with community organizations to address health disparities

Florida

- Targeted topics: Dermatology in T2DM/obesity; Chronic kidney disease in T2DM
- Baseline level: Intermediate—Advanced
- Considerations for future education: Focus on the dermatological complications of metabolic disorders; Highlight the importance of renal function monitoring in T2DM

LEGEND

- **Targeted Topics** (based on Interest/Need)
- **Baseline Level**
- **Considerations for Future Education**

Results & Discussion

Analysis of combined clinician interest and knowledge data revealed opportunities for more efficient educational approaches.[4] By identifying areas where high interest intersects with knowledge gaps, we can develop multifaceted educational programs that maximize learning efficiency. For example, the Southeast region's high interest in women's health combined with knowledge gaps in T2DM/obesity management suggests an opportunity to develop programs that address metabolic disorders within women's health contexts.[3] This strategic matching of educational content to both interest and need directly addresses the critical challenge of time constraints faced by clinicians.[2,4] By delivering education that simultaneously addresses multiple relevant clinical areas while filling specific knowledge gaps, we can provide more immediately applicable learning experiences that offer greater value for busy healthcare providers. This "two-for-one" approach could increase educational efficiency while maintaining high engagement through alignment with clinician interests.

Conclusion

This analysis demonstrates the value of combining clinician interest data with knowledge/confidence assessment metrics to develop more targeted and efficient educational programming. By understanding both what clinicians want to learn and what they need to learn, we can create more engaging and time-efficient educational experiences. Based on our findings, we recommend:

- Development of integrated educational pathways that combine high-interest topics with identified knowledge gaps (e.g., women's health with metabolic disorders, mental health with cardiovascular risk management)
- Implementation of region-specific education that aligns with local interests while addressing documented knowledge deficits
- Creation of "two-for-one" learning opportunities that maximize educational value by addressing multiple, related clinical areas simultaneously

Footnotes

- Analysis limited to static data from a single point in time; continuous assessment needed as healthcare landscape evolves
- Regional sample sizes varied; some regions had limited representation
- AI-assisted data analysis tools were used for regional segmentation
- Further research needed to validate educational impact over time
- Additional and more granular data on file

References

[1] American Diabetes Association. (2021). Standards of Medical Care in Diabetes---2021. Diabetes Care, 44(Supplement 1), S1-S232.

[2] Mitchell, A. J., Vaze, A., & Rao, S. (2009). Clinical diagnosis of depression in primary care: A meta-analysis. The Lancet, 374(9690), 609-619.

[3] Kushner, R. F., & Ryan, D. H. (2014). Assessment and lifestyle management of patients with obesity: Clinical recommendations from systematic reviews. JAMA, 312(9), 943-952.

[4] Kazemian, P., et al. (2019). Evaluation of the Cascade of Diabetes Care in the United States, 2005-2016. JAMA Internal Medicine, 179(10), 1376-1385.